Subgroup Analysis of Participants With HER2 IHC 0 in the ASCENT-07 Study of Sacituzumab Govitecan vs Chemotherapy in HR+/ HER2− Metastatic Breast Cancer

Poster #

Yeon Hee Park[1], Komal Jhaveri[2,3], Carlos Barrios[4], Giuseppe Curigliano[5,6], Hiroji Iwata[7], Javier Cortes[8-10], Delphine Loirat[11], Tomás Pascual[12-15], Zhimin Shao[16,17], Carlos Gallardo Araneda[18], Toshinari Yamashita[19], Marta Tapia[20,21], Pelin Cinar[22], Sivuonthuon Lam[22], Xuehan Ren[22], Brittany Fleming[22], Joyce Kwan[22], Kevin Punie[23], Hope S Rugo[24]

[1]Samsung Medical Center, Sungkyunkwan University School of Medicine, Seoul, Republic of Korea; [2]Memorial Sloan Kettering Cancer Center (MSKCC), New York, NY, USA; [3]Weill Cornell Medical College, New York, NY, USA; [4]Latin American Cooperative Oncology Group (LACOG), Porto Alegre, Brazil; [5]European Institute of Oncology, IRCCS Milano, Milan, Italy; [6]University of Milano, Milan, Italy; [7]Nagoya City University Graduate School of Medical Sciences Medical School, Nagoya, Japan; [8]International Breast Cancer Center (IBCC), Pangaea Oncology, Quiron Group, Barcelona, Spain; [9]Medica Scientia Innovation Research (MEDSIR), Barcelona, Spain; [10]Faculty of Biomedical and Health Sciences, Universidad Europea de Madrid, Madrid, Spain; [11]Institut Curie, Medical Oncology Department and IHU Cancers des Femmes, Paris, France; [12]Cancer Institute and Blood Disorders, Hospital Clínic de Barcelona, Barcelona, Spain; [13]Translational Genomics and Targeted Therapies in Solid Tumors Group, August Pi i Sunyer Biomedical Research Institute (IDIBAPS), Barcelona, Spain; [14]Faculty of Medicine, Universitat de Barcelona, Barcelona, Spain; [15]SOLTI Cancer Research Group, Barcelona, Spain; [16]Fudan University Shanghai Cancer Center, Shanghai, China; [17]Shangai Medical College, Fudan University, Shanghai, China; [18]Bradford Hill Clinical Research Center, Santiago, Chile; [19]Kanagawa Cancer Center, Yokohama, Japan; [20]Hospital Clínico Universitario of Valencia, Valencia, Spain; [21]Biomedical Research Institute INCLIVA, Valencia, Spain; [22]Gilead Sciences, Inc., Foster City, CA, USA; [23]Department of Medical Oncology, Ziekenhuisaan de Stroom, Antwerp, Belgium; [24]City of Hope Comprehensive Cancer Center Duarte, Duarte, CA, USA

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

Conclusions

- In this exploratory analysis of participants with hormone receptor positive (HR+)/human epidermal growth factor receptor 2 (HER2) immunohistochemistry (IHC) 0 metastatic breast cancer (mBC) from the ASCENT-07 study:
 - Sacituzumab govitecan (SG) showed numerical improvement in progression-free survival (PFS) vs treatment of physician's choice (TPC) by both blinded independent central review (BICR) and investigator assessments
 - SG showed an early trend in improvement of overall survival (OS) over TPC, although these data were not mature (28% maturity)
 - Objective response rate (ORR) was similar between treatment groups, and median duration of response (DOR) was longer with SG vs TPC
 - The safety profile of SG was manageable and consistent with the overall study population[5]
- These findings support further evaluation of SG in patients with HR+/HER2 IHC 0 mBC who are candidates for first chemotherapy

Plain Language Summary

- The ASCENT-07 study showed that, people with advanced hormone receptor positive/human epidermal growth factor receptor 2 (HER2) negative breast cancer previously treated with hormone therapy and receiving sacituzumab govitecan (SG) had a similar risk of their cancer growing or spreading as those receiving chemotherapy
- This analysis focused on participants from ASCENT-07 whose tumors were HER2 IHC 0, meaning their cancer cells showed no detectable HER2 protein. These patients have few treatment options
- When reviewed by independent radiologists, participants receiving SG lived slightly longer without their cancer growing or spreading (median [the middle value of a dataset] 9.2 months) than those receiving chemotherapy (median 8.1 months). Local researchers reported similar findings (8.5 vs 6.1 months)
- Participants treated with SG appeared to live longer overall than those treated with chemotherapy
- Both treatments shrank tumors at similar rates, but responses lasted longer with SG
- The safety profile of SG was similar to that observed previously and can be managed by medical professionals
- These findings suggest that it may be valuable to further study SG in patients with advanced hormone receptor positive/HER2 IHC 0 breast cancer who are eligible to receive chemotherapy

References: **1.** Wolff AC, et al. *J Clin Oncol*. 2023;41:3867-72. **2.** Bagegni NA, et al. *Cancers (Basel)*. 2025;17:4021. **3.** TRODELVY® (sacituzumab govitecan-hziy) [prescribing information]. Foster City, CA: Gilead Sciences, Inc., March 2025. **4.** Rugo H, et al. *Lancet*. 2023;402:1423-33. **5.** Jhaveri KL, et al. *Clin Cancer Res*. 2026;32(4_Supp):GS1-09.

Acknowledgments: We extend our thanks to the patients, their families, and all participating investigators. This study was funded by Gilead Sciences, Inc. Writing and editorial assistance were provided by Priya Talluri, MSc, of Parexel, and funded by Gilead Sciences, Inc.

Disclosures: **YHP** reports consulting or advisory roles with Pfizer, Roche, Eisai, Daiichi Sankyo, AstraZeneca, MSD, Novartis, Lilly, Menarini, Gilead Sciences, Boryung, Helsinn Therapeutics, Bristol Myers Squibb, AimedBio, Voronoi Health Analytics, and Jazz Pharmaceuticals; travel, accommodation, and expenses from Gilead Sciences, Jazz Pharmaceuticals, Roche, Novartis, Pfizer, Daiichi Sankyo, and Helsinn Therapeutics; honoraria from Novartis, Pfizer, Roche, Lilly, AstraZeneca, MSD, Eisai, Daiichi Sankyo, Helsinn Healthcare, Gilead Sciences, Bristol Myers Squibb, AimedBio, Voronoi Health Analytics, and BeOne Medicines; research funding from AstraZeneca, Pfizer, Roche, Gencurix, NGeneBio, Inocras, Boryung, Daewoong Pharmaceutical, Kirin Pharmaceuticals, Gilead Sciences, Gencurix, and Inocras.

Correspondence: Yeon Hee Park, yeonh.park@samsung.com

Introduction

- Despite new treatments for HER2-low (IHC 1+ or 2+/in situ hybridization−) mBC, patients with IHC 0 (absent or incomplete membrane staining in ≤ 10% of tumor cells, including null and HER2-ultralow) have limited options, particularly in the advanced/metastatic setting[1,2]
- SG is approved for HR+/HER2− mBC after prior endocrine therapy (ET) and ≥ 2 systemic therapies in the metastatic setting after demonstrating a statistically significant and clinically meaningful improvement in PFS and OS vs chemotherapy in the TROPiCS-02 study[3,4]
- ASCENT-07 is a randomized phase 3 study of SG vs TPC in adults with HR+/HER2−, locally advanced unresectable or mBC who had received prior ET and were eligible for first chemotherapy[5]
 - The primary endpoint (PFS by BICR) was not met (median PFS, 8.3 months for both SG and TPC; HR, 0.85; 95% CI, 0.69-1.05; *P* = 0.130); OS data were not yet mature, with follow-up ongoing[5]
- This analysis reports exploratory outcomes in participants with HER2 IHC 0 expression status from ASCENT-07

Methods

- Study design, eligibility criteria, and endpoints are shown in **Figure 1**
- Descriptive analyses of participants with HER2 IHC 0 were conducted for PFS by BICR (primary endpoint), as well as PFS by investigator, OS, ORR and DOR by BICR, and safety (secondary endpoints)
- The September 2025 data cutoff is the same cutoff used for the primary analysis of the overall study population
- HER2 status was tested locally, not centrally

Figure 1. ASCENT-07 Study Design

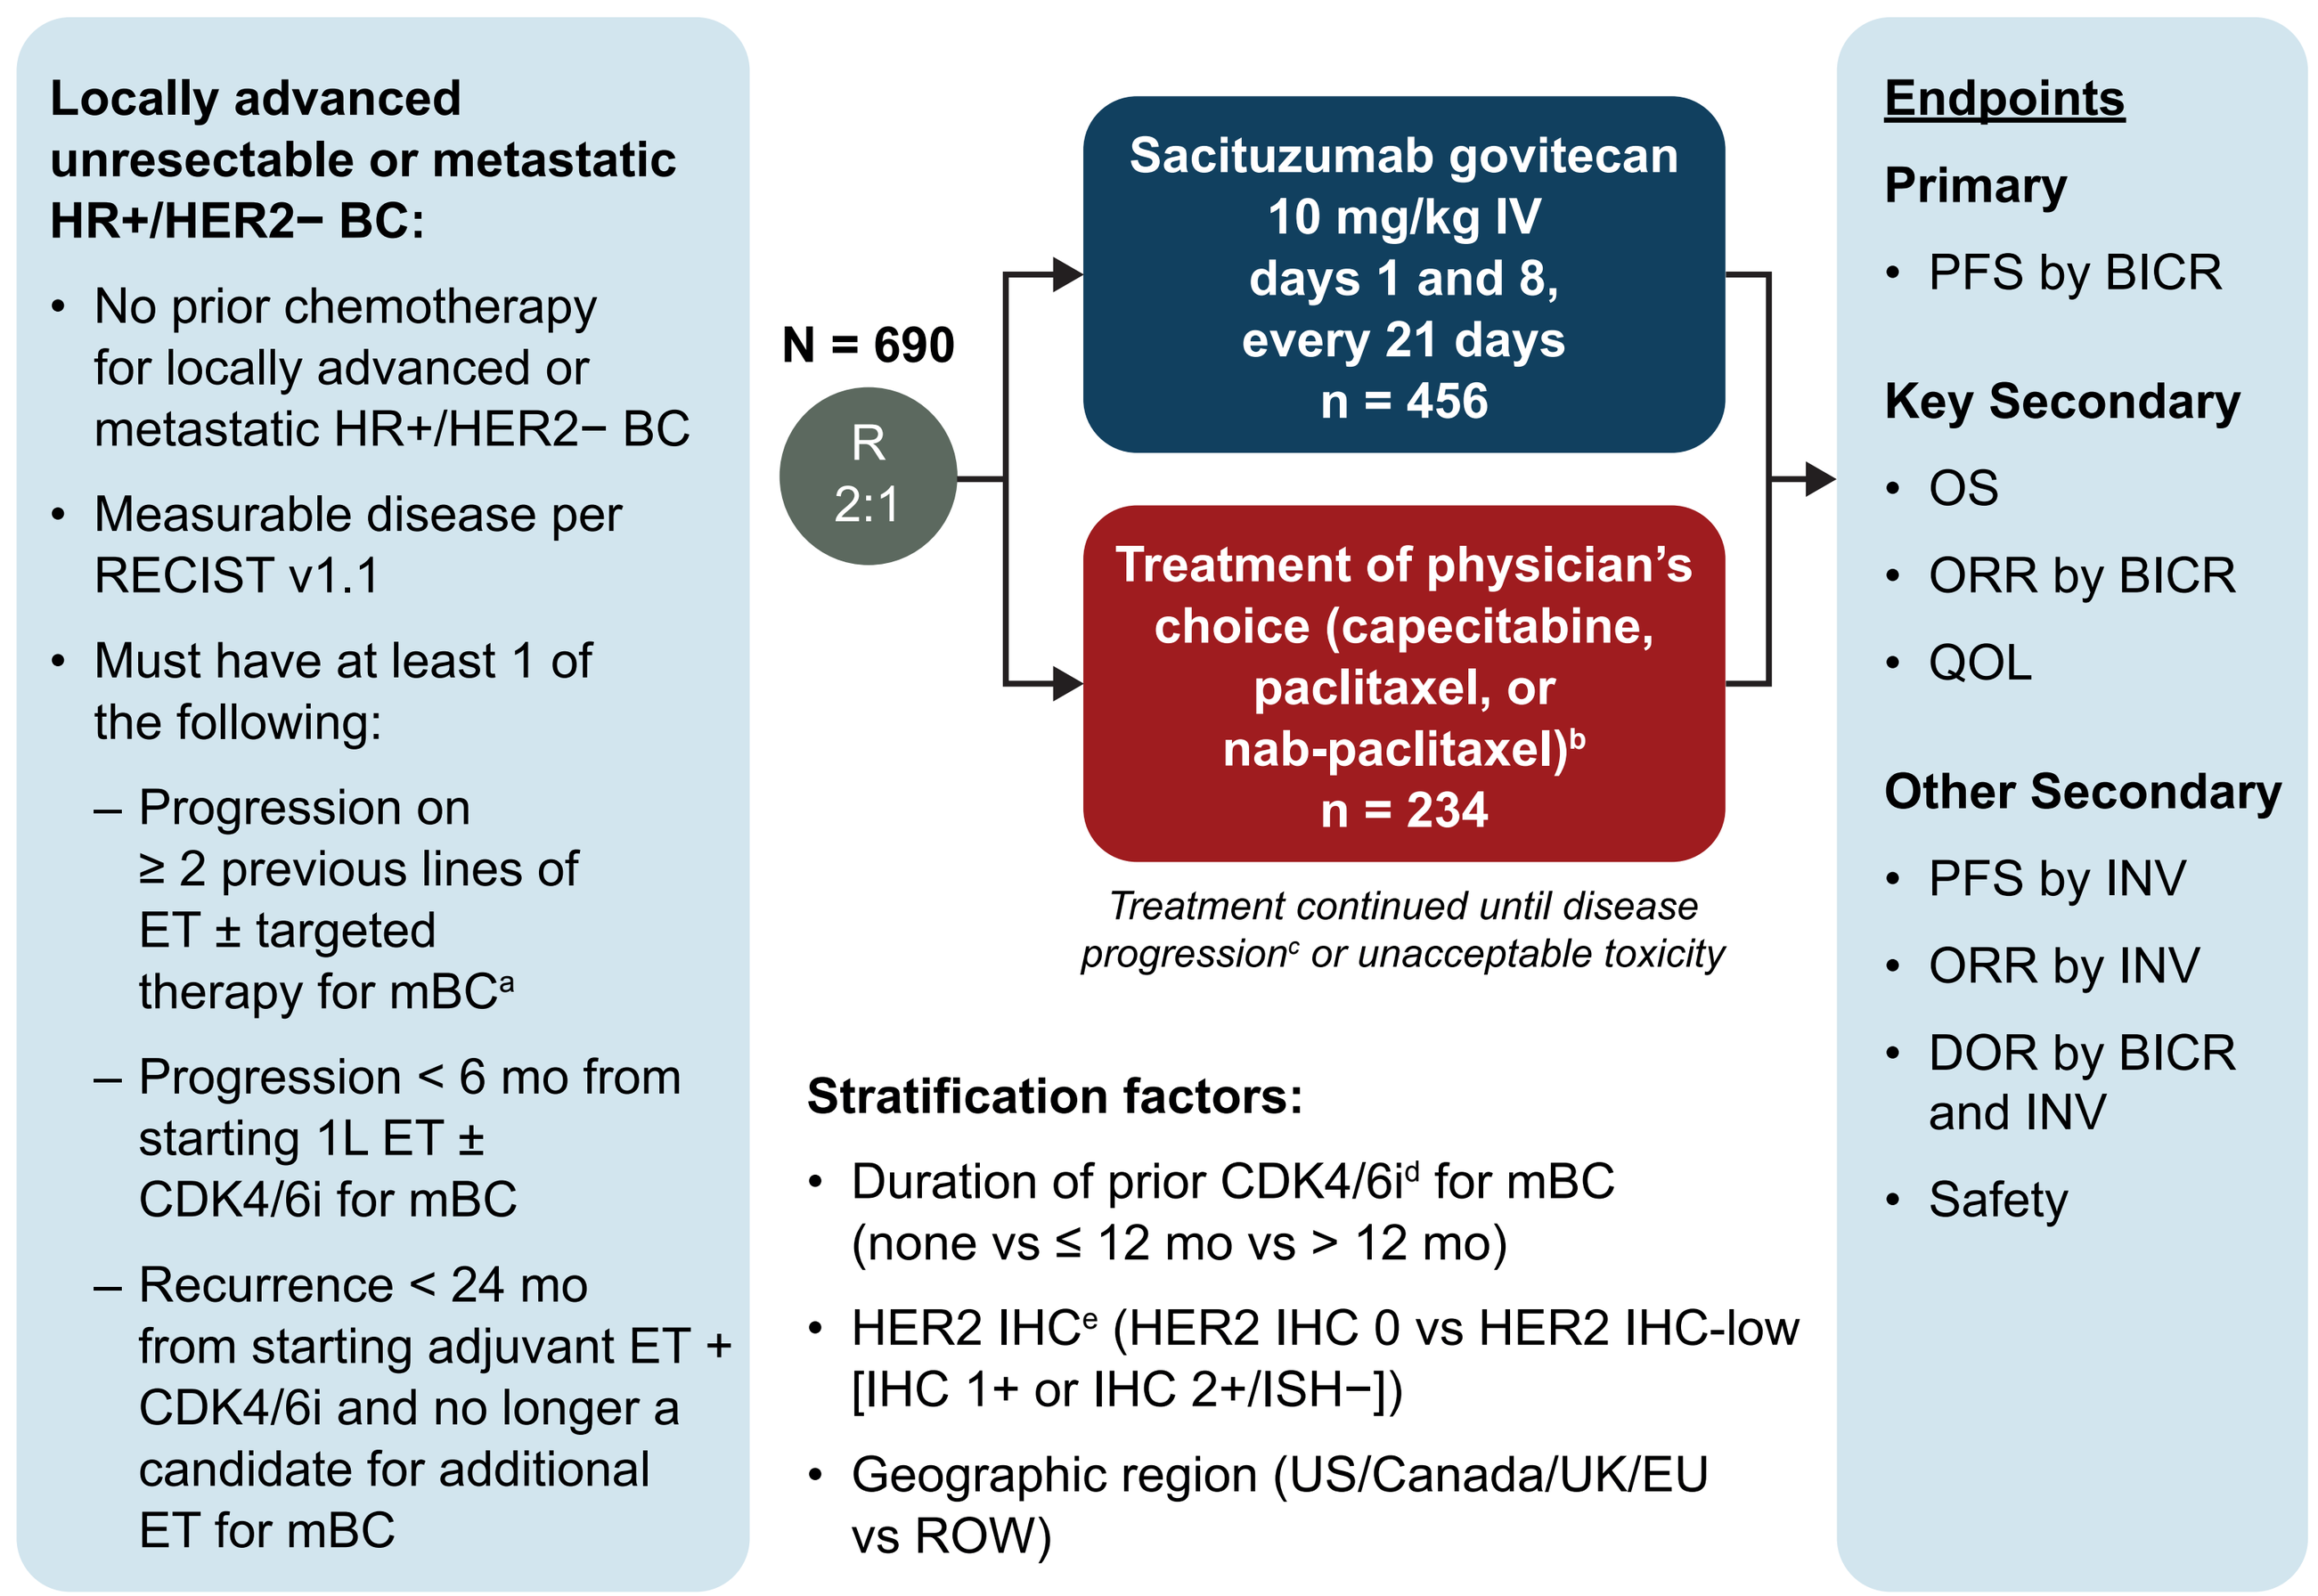
Locally advanced unresectable or metastatic HR+/HER2− BC:
• No prior chemotherapy for locally advanced or metastatic HR+/HER2− BC
• Measurable disease per RECIST v1.1
• Must have at least 1 of the following:
– Progression on ≥ 2 previous lines of ET ± targeted therapy for mBC[a]
– Progression < 6 mo from starting 1L ET ± CDK4/6i for mBC
– Recurrence < 24 mo from starting adjuvant ET + CDK4/6i and no longer a candidate for additional ET for mBC
N = 690
R 2:1
Sacituzumab govitecan 10 mg/kg IV days 1 and 8, every 21 days n = 456
Treatment of physician's choice (capecitabine, paclitaxel, or nab-paclitaxel)[b] n = 234
Treatment continued until disease progression[c] or unacceptable toxicity
Stratification factors:
• Duration of prior CDK4/6i[d] for mBC (none vs ≤ 12 mo vs > 12 mo)
• HER2 IHC[e] (HER2 IHC 0 vs HER2 IHC-low [IHC 1+ or IHC 2+/ISH−])
• Geographic region (US/Canada/UK/EU vs ROW)
Endpoints
Primary
• PFS by BICR
Key Secondary
• OS
• ORR by BICR
• QOL
Other Secondary
• PFS by INV
• ORR by INV
• DOR by BICR and INV
• Safety

ClinicalTrials.gov identifier: NCT05840211.
[a]Disease recurrence while on the first 24 months of starting adjuvant ET will be considered a line of therapy; these participants will only require 1 line of ET in the metastatic setting. [b]Paclitaxel 80 mg/m² or nab-paclitaxel 100 mg/m² IV on days 1, 8, and 15 of 28-day cycles, or capecitabine oral 1000 or 1250 mg/m² twice daily for first 2 weeks of 21-day cycles. [c]Per RECIST v1.1. [d]Enrollment of CDK4/6i-naive participants was capped at 10%. [e]Per local testing.
1L, first-line; BC, breast cancer; BICR, blinded independent central review; CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; DOR, duration of response; ET, endocrine therapy; EU, European Union; HER2−, human epidermal growth factor receptor 2 negative; HR+, hormone receptor positive; IHC, immunohistochemistry; INV, investigator assessment; ISH, in situ hybridization; IV, intravenously; mBC, metastatic breast cancer; mo, months; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; QOL, quality of life; R, randomized; RECIST v1.1, Response Evaluation Criteria in Solid Tumors, version 1.1; ROW, rest of the world; UK, United Kingdom; US, United States.

Results

- Of 690 participants, 292 (42%) were HER2 IHC 0 (SG: 192; TPC: 100)
- Baseline demographics and characteristics were similar to the overall population[5]
- Median age was 58 years; 88% of participants in the SG group and 86% in the TPC group had visceral disease, and 70% and 73%, respectively, had liver metastasis (**Table 1**)
- Most participants received prior ET + cyclin-dependent kinase 4/6 inhibitor in the metastatic setting (SG: 92%; TPC: 91%)
- At 15.4-month median follow-up, SG showed numerical improvement vs TPC in PFS by BICR: 9.2 vs 8.1 months (HR, 0.75; 95% CI, 0.55-1.02; nominal *P* = 0.0601; **Figure 2**) and 8.5 vs 6.1 months by investigator (HR, 0.66; 95% CI, 0.50-0.86; nominal *P* = 0.0024)
- Although OS data were not mature (28% maturity in IHC 0 subgroup), an early improvement trend favoring SG was observed (**Figure 3**)
- 58% of participants in the TPC group received an antibody-drug conjugate following treatment discontinuation

Table 1. Demographics and Baseline Characteristics in IHC 0 Subgroup

Characteristic	SG (n = 192)	TPC (n = 100)
Female, n (%)	192 (100)	100 (100)
Median age, years (range)	59 (29-86)	56 (27-78)
≥ 65 years, n (%)	52 (27)	29 (29)
Geographic region, n (%)		
US/Canada/UK/EU	75 (39)	39 (39)
Rest of the world	117 (61)	61 (61)
Race,[a] n (%)		
White	113 (59)	49 (49)
Asian	57 (30)	38 (38)
Black	4 (2)	1 (1)
Other/not specified	18 (9)	12 (12)
ECOG PS at baseline,[b] n (%)		
0	117 (61)	63 (63)
1	75 (39)	37 (37)
ER/PR status,[c] n (%)		
ER+ and PR+	127 (66)	68 (68)
ER+ and PR−	61 (32)	30 (30)
ER− and PR+	1 (< 1)	2 (2)
Primary endocrine resistance,[d] n (%)	58 (30)	26 (26)
Time from metastatic diagnosis to randomization, median (range) months	24.2 (12.0-40.2)	26.7 (11.9-45.8)
De novo metastatic disease at diagnosis, n (%)	49 (26)	22 (22)
Visceral disease, n (%)	169 (88)	86 (86)
Liver metastasis, n (%)	135 (70)	73 (73)
Brain metastasis, n (%)	6 (3)	9 (9)
Bone-only disease, n (%)	9 (5)	7 (7)

[a]As reported by the participants; Other/not specified includes American Indian or Alaska Native, other races, and not provided/collection not permitted. [b]Scores range from 0 to 5, with higher scores indicating greater disability. [c]ER/PR status was missing for 3 participants. [d]Defined as relapse that had occurred during the first 2 years of adjuvant endocrine therapy or progressive disease that had occurred during the first 6 months of first-line endocrine therapy for metastatic breast cancer.
ECOG PS, Eastern Cooperative Oncology Group performance status; ER, estrogen receptor; EU, European Union; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; PR, progesterone receptor; SG, sacituzumab govitecan; TPC, treatment of physician's choice; UK, United Kingdom; US, United States.

Figure 2. Progression-Free Survival by BICR in IHC 0 Subgroup

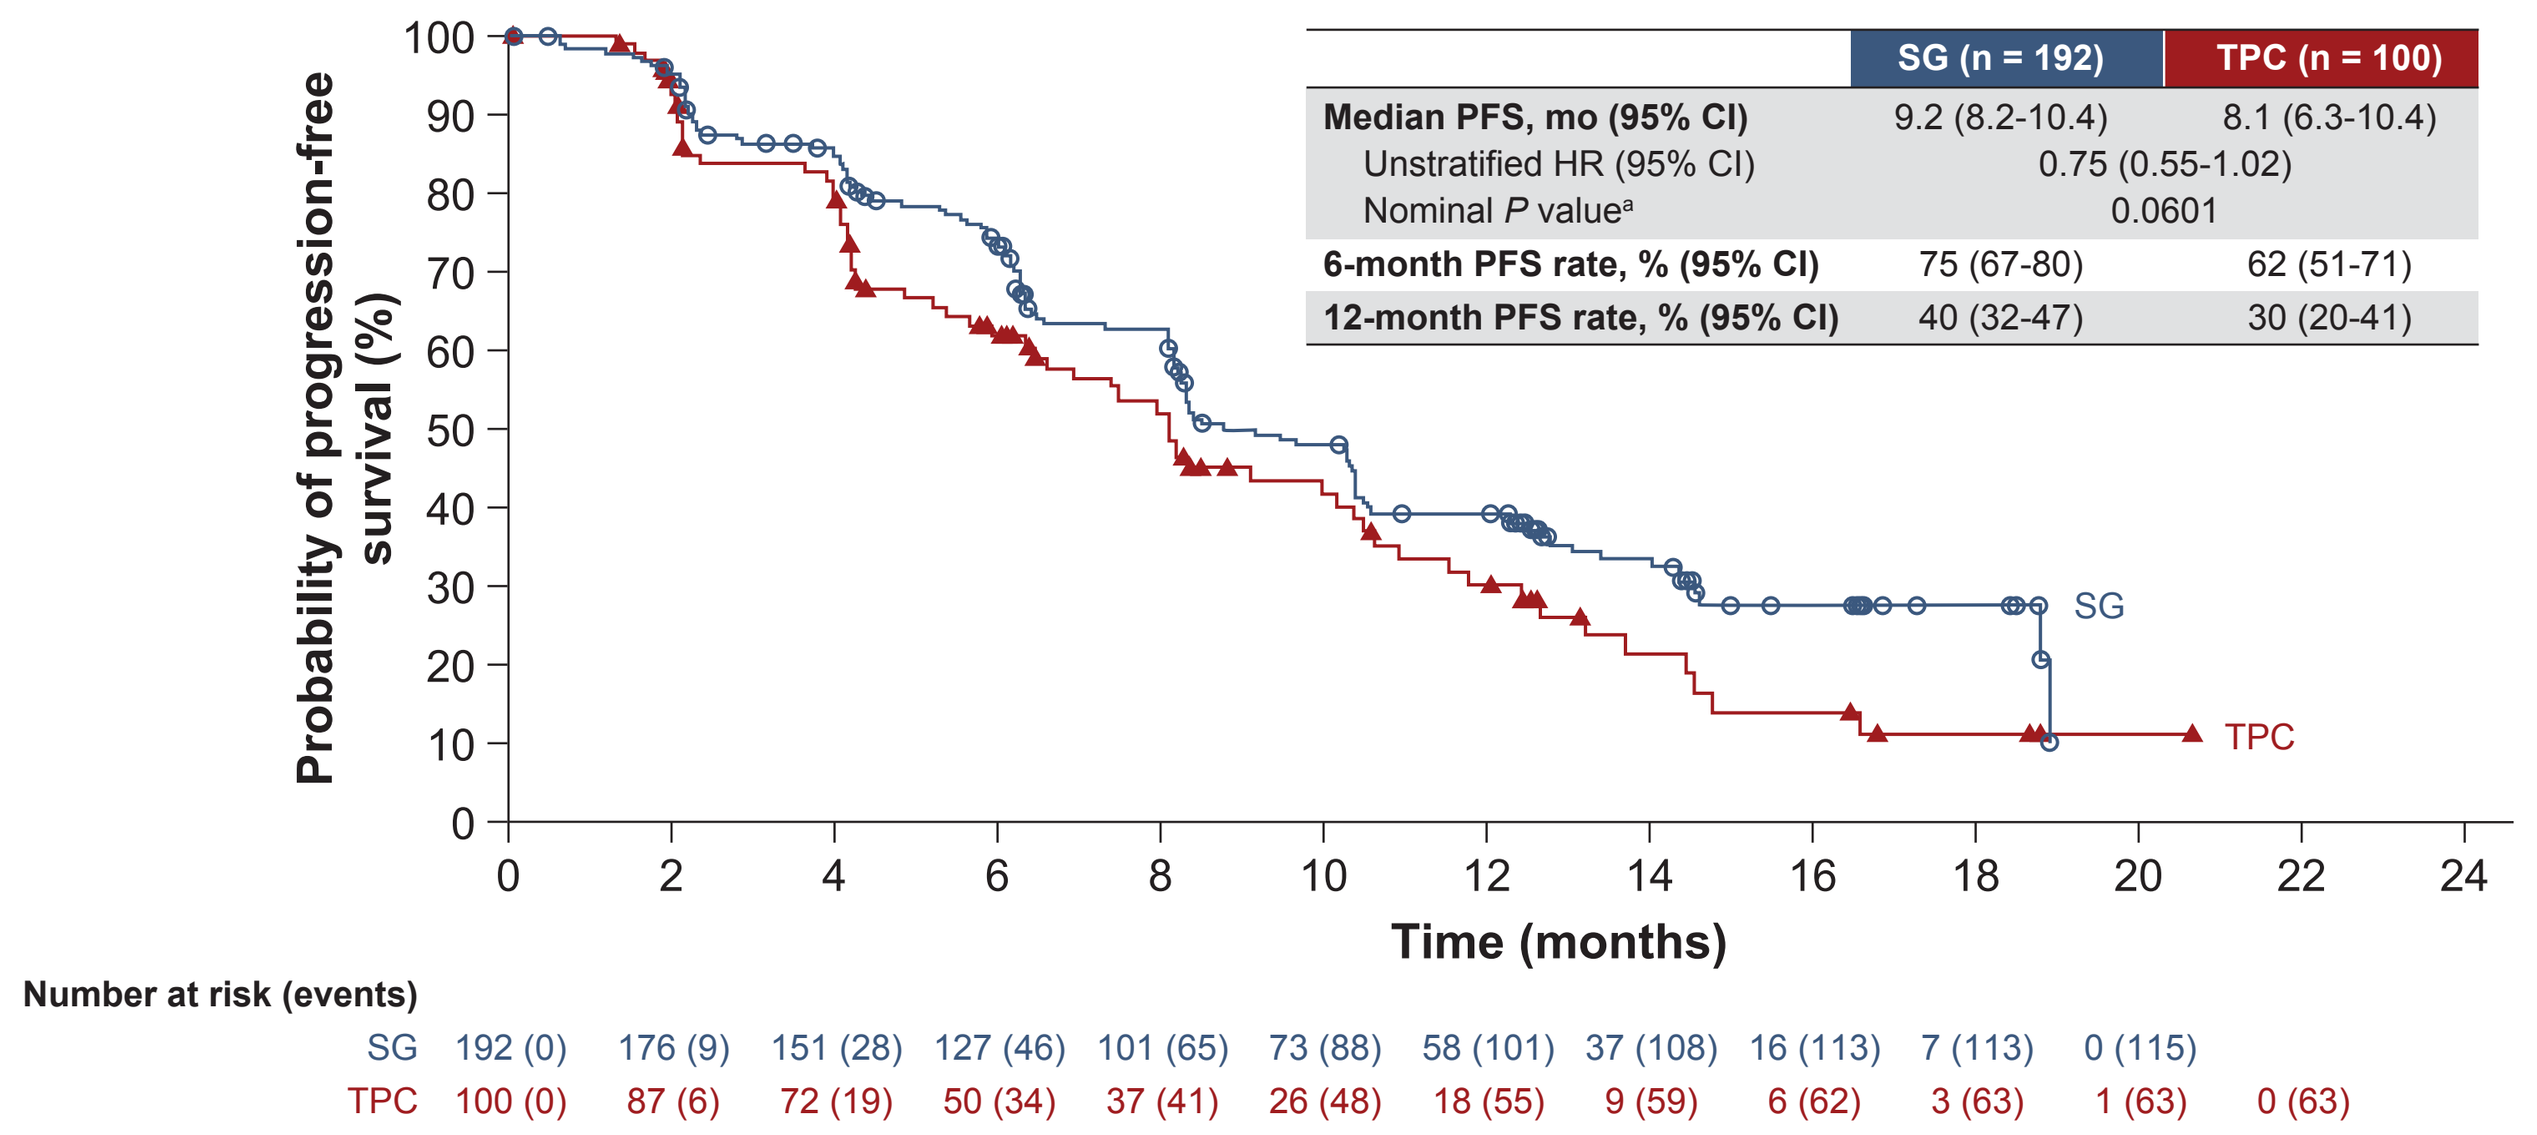
Probability of progression-free survival (%)
Time (months)
SG
TPC
Number at risk (events)
SG 192 (0) 176 (9) 151 (28) 127 (46) 101 (65) 73 (88) 58 (101) 37 (108) 16 (113) 7 (113) 0 (115)
TPC 100 (0) 87 (6) 72 (19) 50 (34) 37 (41) 26 (48) 18 (55) 9 (59) 6 (62) 3 (63) 1 (63) 0 (63)

	SG (n = 192)	TPC (n = 100)
Median PFS, mo (95% CI)	9.2 (8.2-10.4)	8.1 (6.3-10.4)
Unstratified HR (95% CI)	0.75 (0.55-1.02)	
Nominal *P* value[a]	0.0601	
6-month PFS rate, % (95% CI)	75 (67-80)	62 (51-71)
12-month PFS rate, % (95% CI)	40 (32-47)	30 (20-41)

Median follow-up was 15.4 months. [a]Two-sided P value from unstratified log-rank test.
BICR, blinded independent central review; HR, hazard ratio; IHC, immunohistochemistry; mo, months; PFS, progression-free survival; SG, sacituzumab govitecan; TPC, treatment of physician's choice.

Figure 3. Overall Survival in IHC 0 Subgroup (28% Maturity)

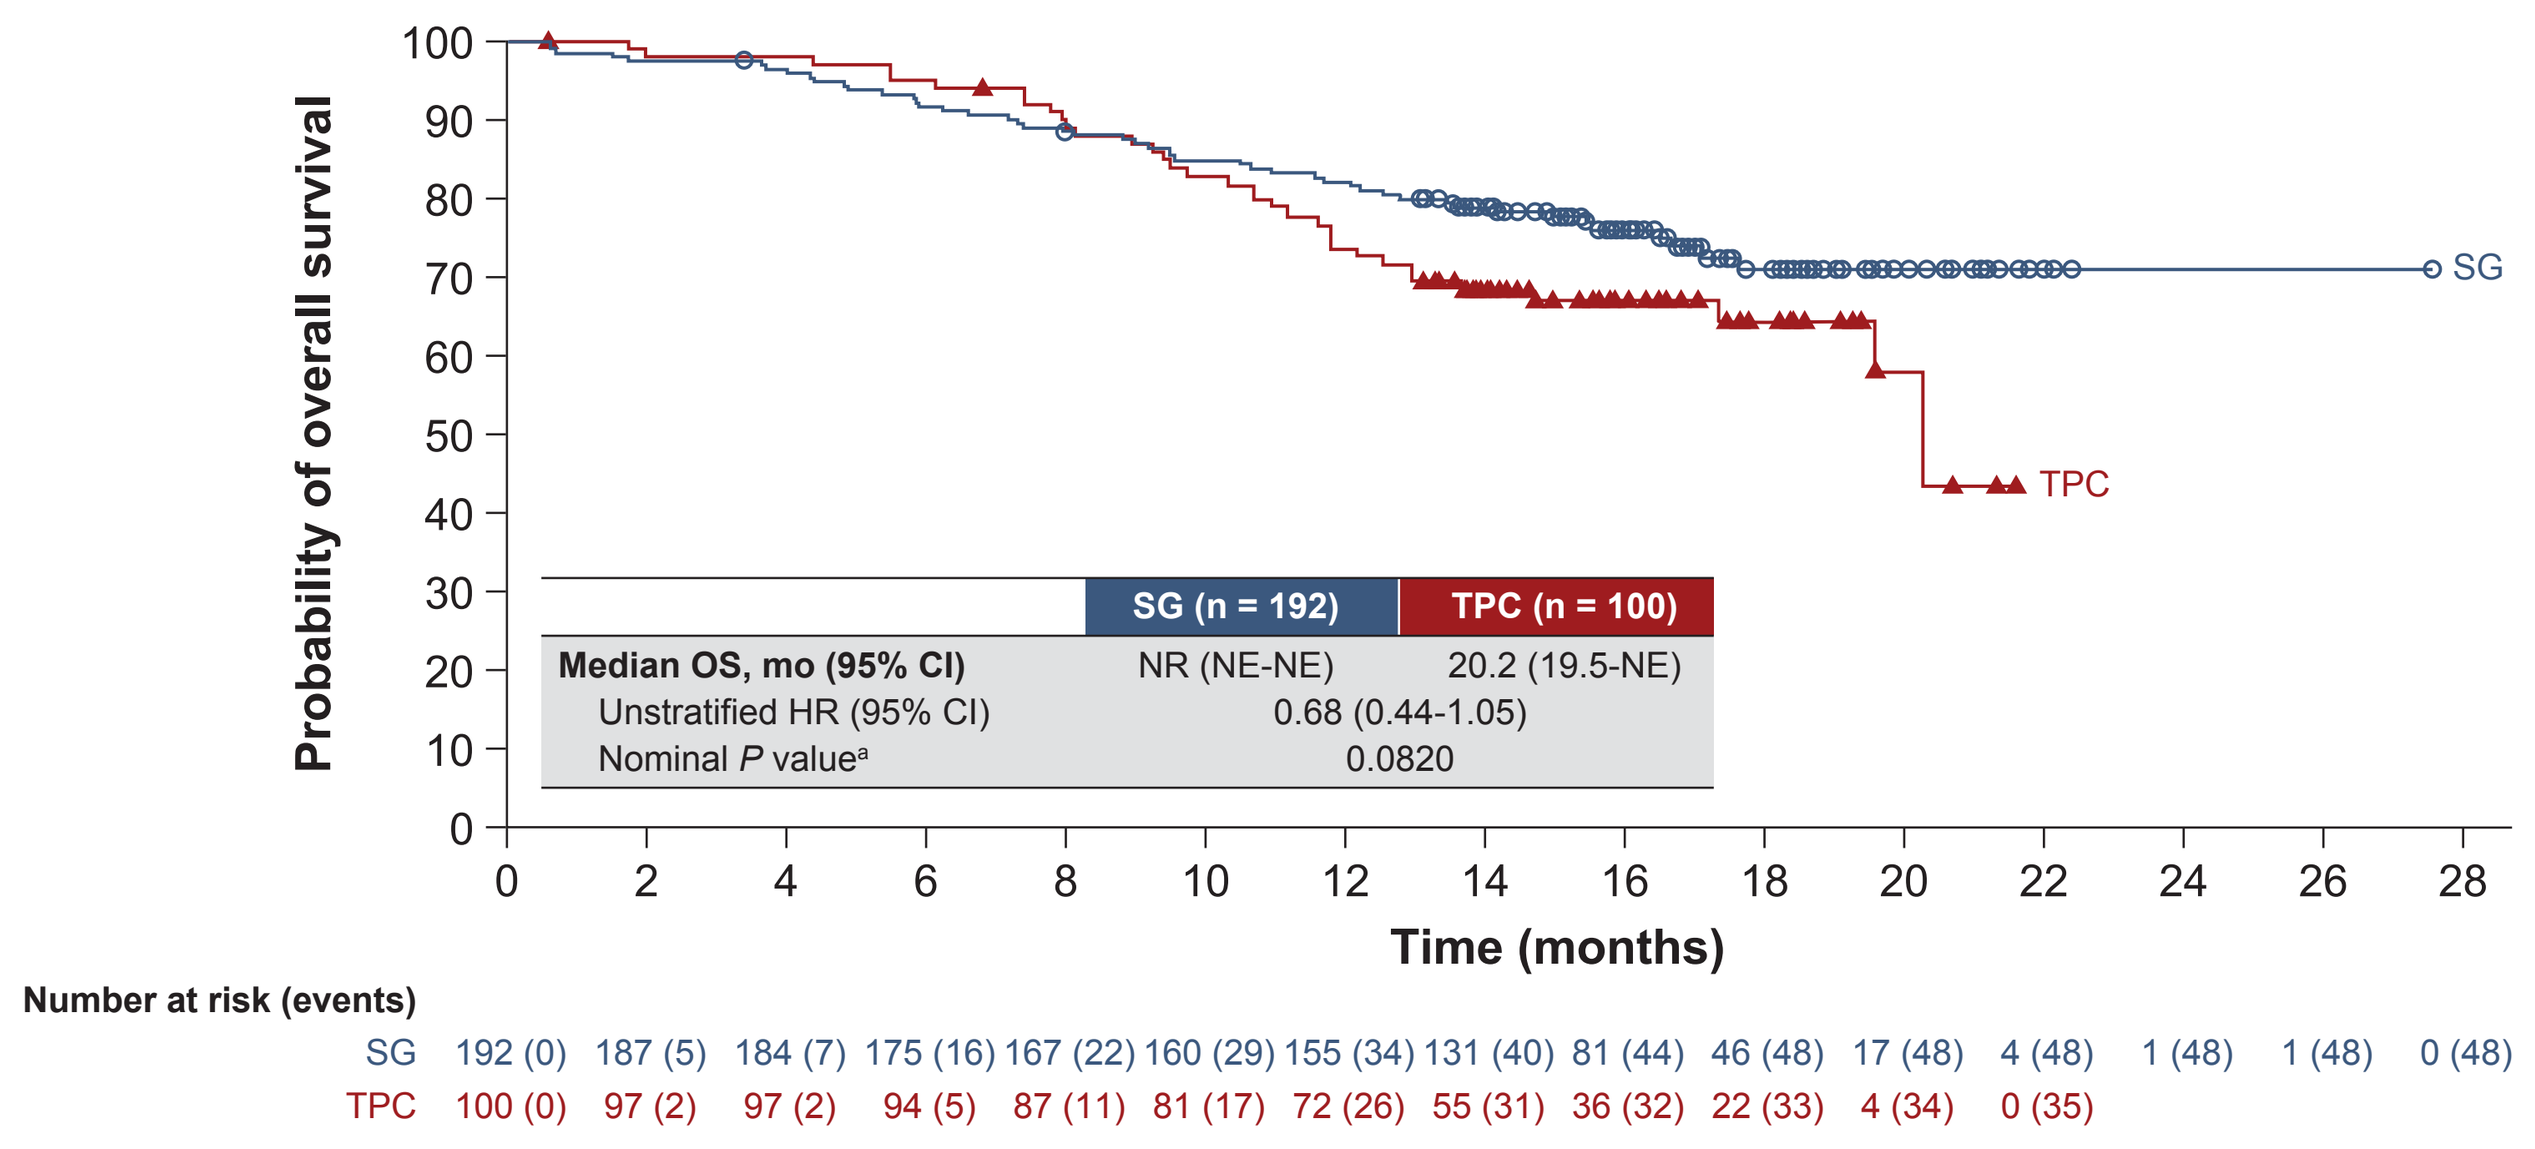
Probability of overall survival
Time (months)
SG
TPC
Number at risk (events)
SG 192 (0) 187 (5) 184 (7) 175 (16) 167 (22) 160 (29) 155 (34) 131 (40) 81 (44) 46 (48) 17 (48) 4 (48) 1 (48) 1 (48) 0 (48)
TPC 100 (0) 97 (2) 97 (2) 94 (5) 87 (11) 81 (17) 72 (26) 55 (31) 36 (32) 22 (33) 4 (34) 0 (35)

	SG (n = 192)	TPC (n = 100)
Median OS, mo (95% CI)	NR (NE-NE)	20.2 (19.5-NE)
Unstratified HR (95% CI)	0.68 (0.44-1.05)	
Nominal *P* value[a]	0.0820	

[a]The nominal *P* value was reported as a descriptive measure of the observed treatment effect and does not support statistical significance.
HR, hazard ratio; IHC, immunohistochemistry; mo, months; NE, not evaluable; NR, not reached; OS, overall survival; SG, sacituzumab govitecan; TPC, treatment of physician's choice.

- ORR by BICR was similar for SG and TPC, and median DOR by BICR was longer for SG vs TPC (**Figure 4**)

Figure 4. Tumor Responses and Duration of Response by BICR in IHC 0 Subgroup

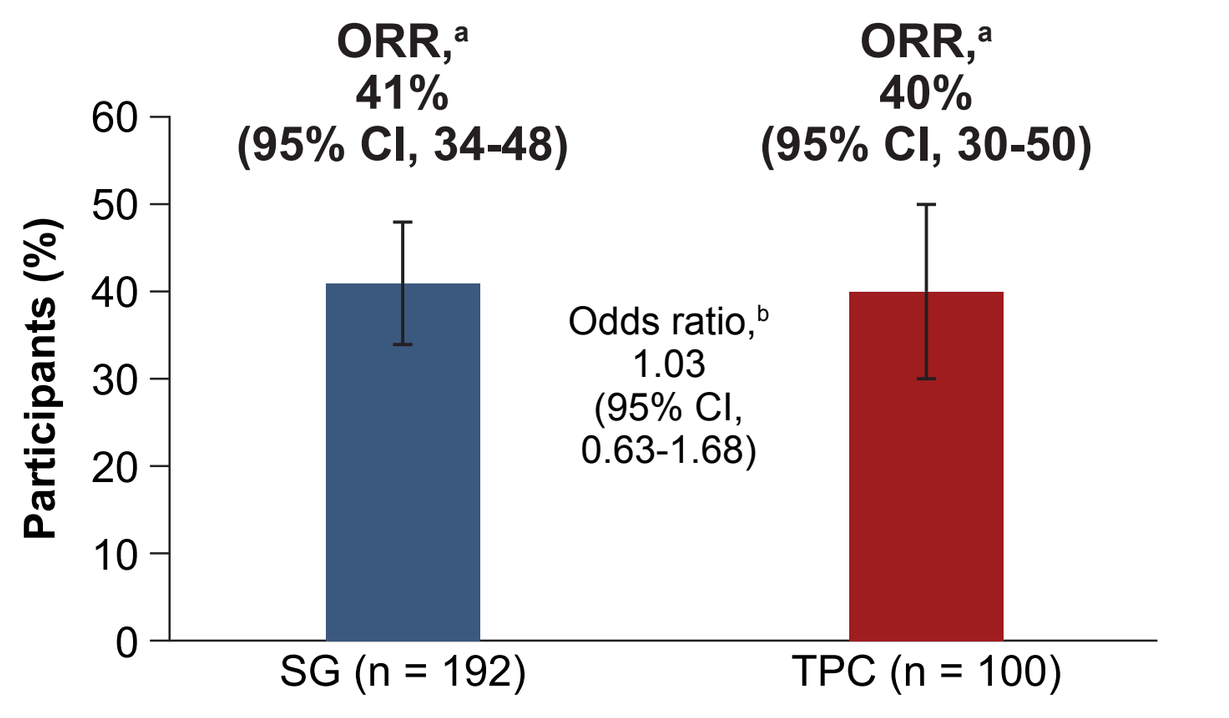
ORR,[a] 41% (95% CI, 34-48)
ORR,[a] 40% (95% CI, 30-50)
Odds ratio,[b] 1.03 (95% CI, 0.63-1.68)
Participants (%)
SG (n = 192)
TPC (n = 100)

Variable	SG (n = 192)	TPC (n = 100)
Clinical benefit rate,[c] % (95% CI)	70 (63-76)	55 (45-65)
Responders	**SG (n =78)**	**TPC (n = 40)**
Median time to response,[d] months (range)	2.2 (1.4-10.5)	3.0 (1.9-10.4)
Median duration of response, months (95% CI)	10.8 (6.4-12.6)	8.3 (5.5-11.2)

[a]ORR is defined as the proportion of participants who achieved a best overall response of complete response/partial response. [b]Odds ratio and 95% CI are calculated from unstratified Mantel-Haenszel estimates without any adjustment. [c]Clinical benefit rate is defined as the proportion of participants who achieved best overall response of CR/PR or durable SD with duration ≥ 6 months; the 95% CI is based on Clopper-Pearson method. [d]Time to response (months) = (date of first documented complete or partial response - date of randomization + 1)/30.4375.
BICR, blinded independent central review; CR, complete response; IHC, immunohistochemistry; ORR, objective response rate; PR, partial response; SD, stable disease; SG, sacituzumab govitecan; TPC, treatment of physician's choice.

- Although more participants had grade ≥ 3 treatment-emergent adverse events (TEAEs) with SG vs TPC, the rate of treatment discontinuation was lower with SG (**Table 2**)

Table 2. Exposure and Safety Summary in IHC 0 Subgroup

AEs, n (%)	SG (n = 190)	TPC (n = 100)
Any TEAE	189 (99)	97 (97)
Treatment related	189 (99)	91 (91)
Grade ≥ 3 TEAEs	139 (73)	46 (46)
Treatment related	131 (69)	35 (35)
Treatment-emergent SAEs	51 (27)	13 (13)
Treatment related	37 (20)	3 (3)
TEAEs leading to treatment discontinuation	4 (2)	7 (7)
TEAEs leading to dose interruption	146 (77)	44 (44)
TEAEs leading to dose reduction	83 (44)	35 (35)
TEAEs leading to death	4 (2)	3 (3)
Treatment related	4 (2)	1 (1)
Median (range) duration of treatment, months	8.3 (0.0-21.4)	6.1 (0.4-21.1)
Median (range) relative dose intensity,[a] %	83.7 (33.1-127.3)	93.4 (47.5-108.4)

TEAEs began on or after the first dose date of study drug up to 30 days after the last dose date of the study drug, or the day prior to the initiation of subsequent anticancer therapy, whichever occurred first.
[a]Relative dose intensity calculated as (total amount of study drug administered/total amount of actual study drug planned by protocol)*100.
AE, adverse event; IHC, immunohistochemistry; SAE, serious adverse event; SG, sacituzumab govitecan; TEAE, treatment-emergent adverse event; TPC, treatment of physician's choice.

- The most common grade ≥ 3 TEAEs were neutropenia (SG: 54%; TPC: 21%), leukopenia (12%; 7%), and anemia (11%; 5%) (**Figure 5**)
- The safety profile was consistent with the overall population and prior studies[4,5]

Figure 5. Most Common (Occurring in ≥ 20%) Treatment-Emergent Adverse Events in IHC 0 Subgroup

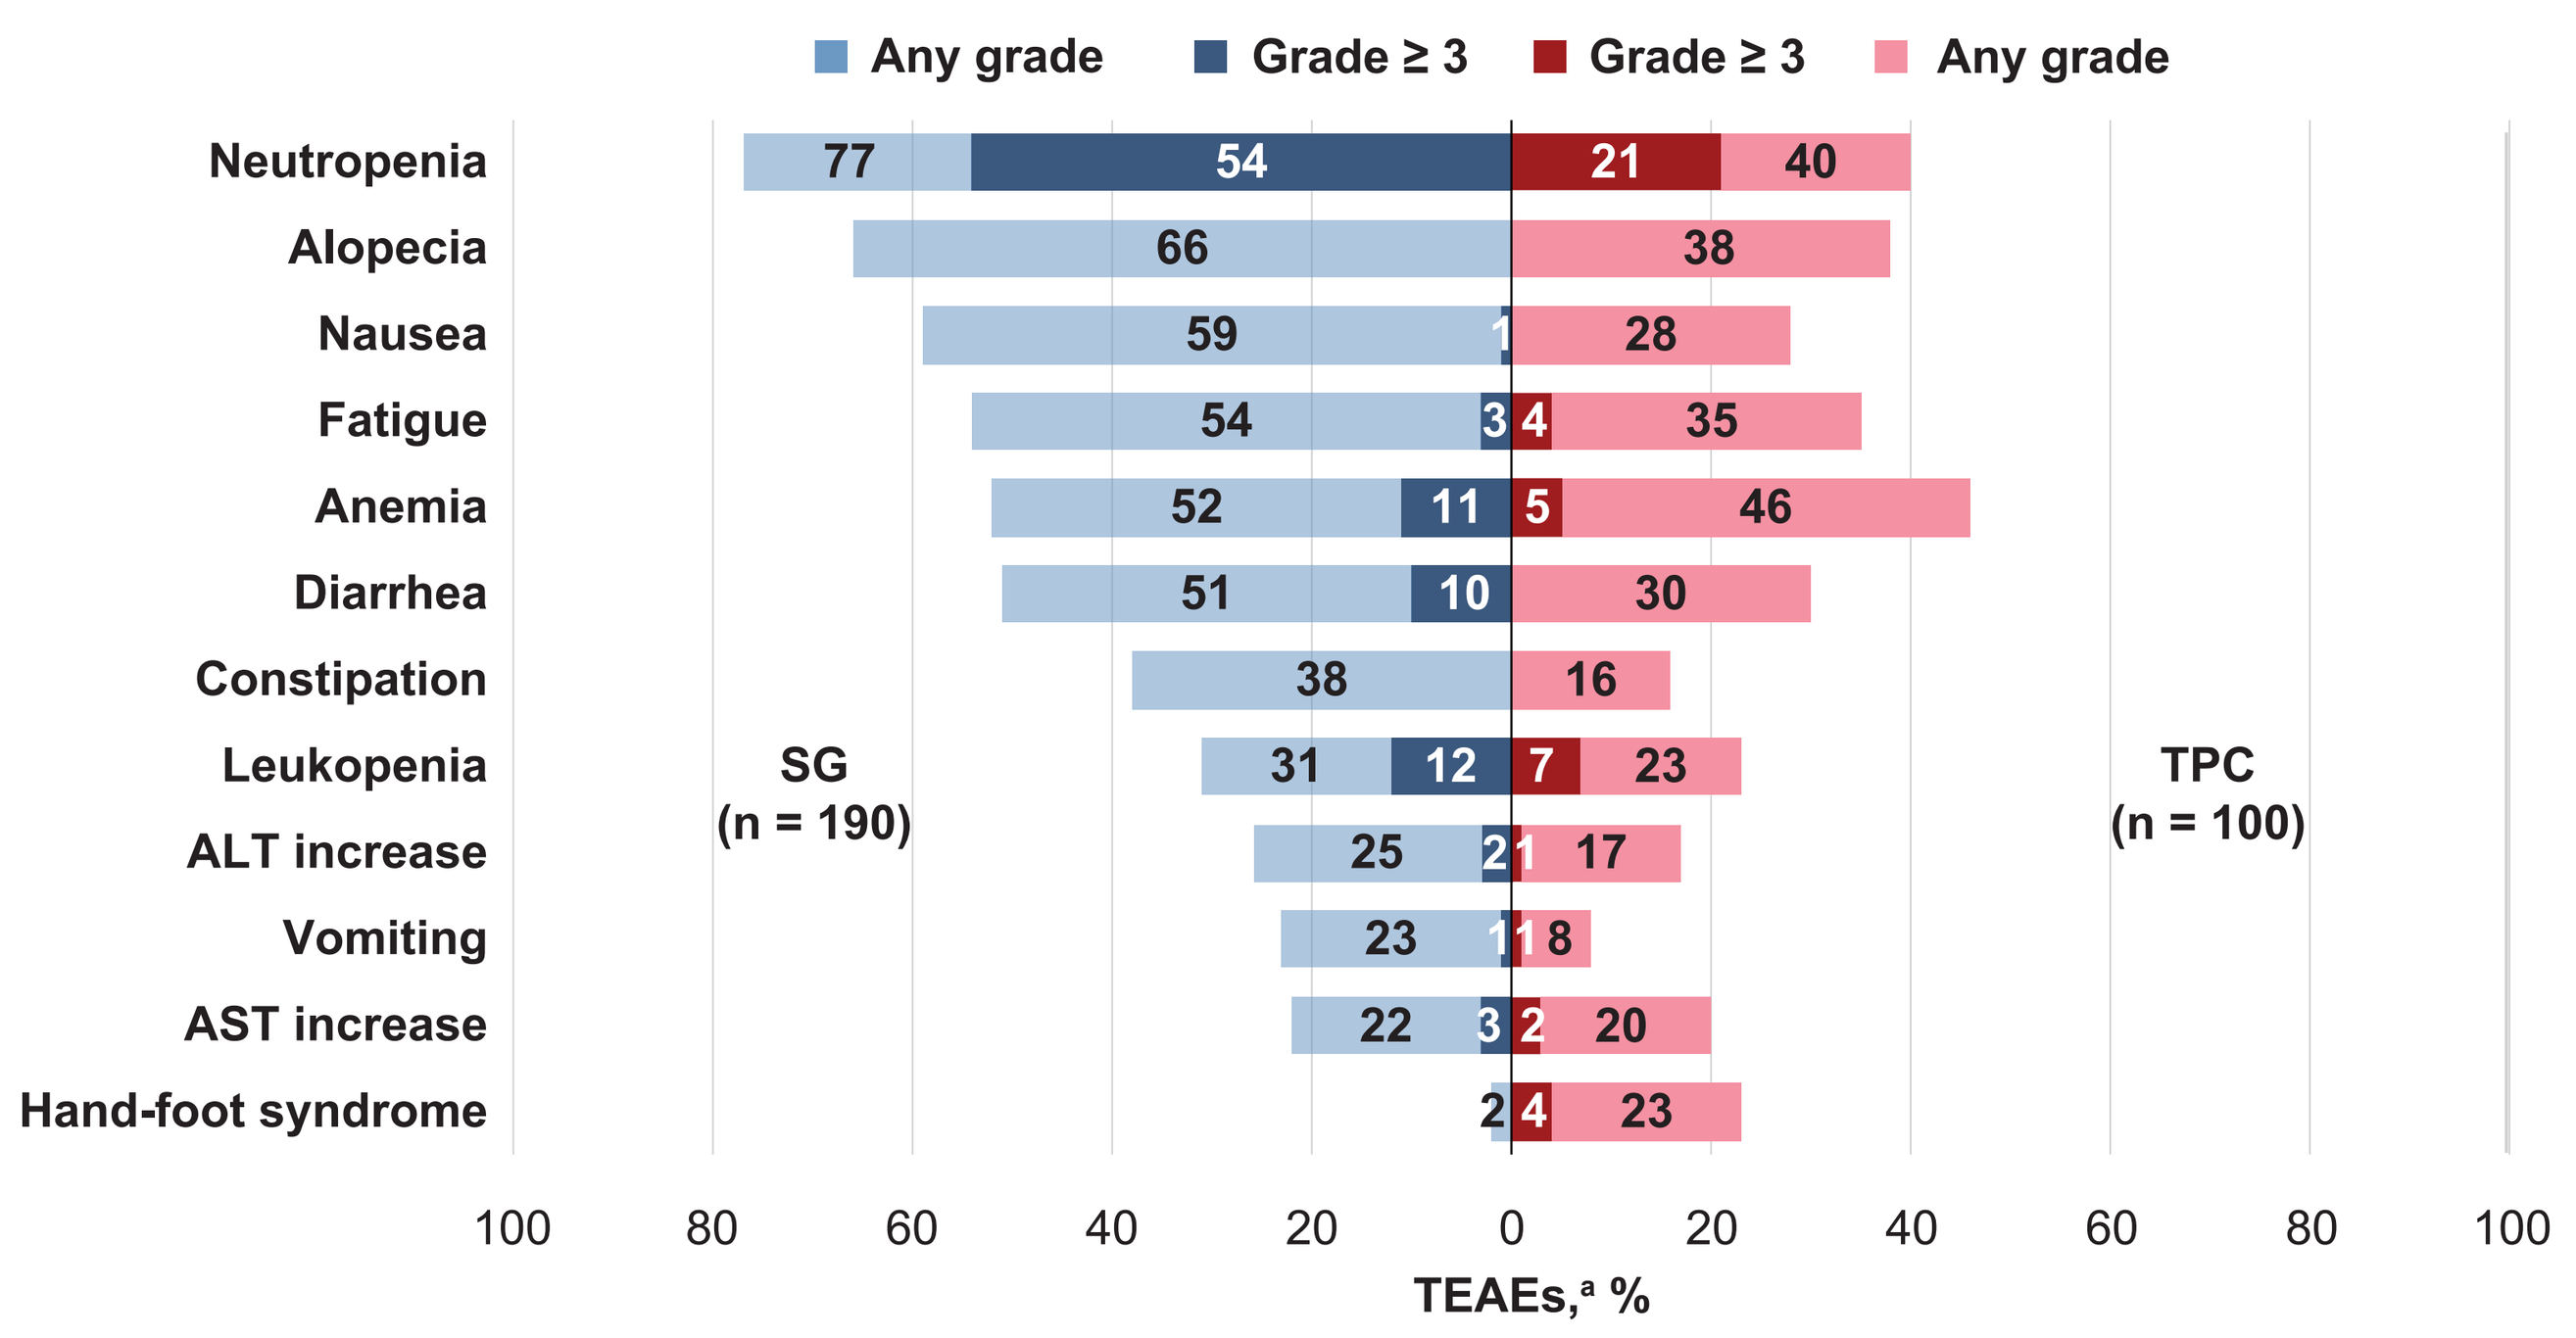
Any grade
Grade ≥ 3
Grade ≥ 3
Any grade
Neutropenia 77 54 21 40
Alopecia 66 38
Nausea 59 1 28
Fatigue 54 3 4 35
Anemia 52 11 5 46
Diarrhea 51 10 30
Constipation 38 16
Leukopenia 31 12 7 23
ALT increase 25 2 1 17
Vomiting 23 1 1 8
AST increase 22 3 2 20
Hand-foot syndrome 2 4 23
SG (n = 190)
TPC (n = 100)
TEAEs,[a] %

TEAEs began on or after the first dose date of study drug up to 30 days after the last dose date of the study drug or the day before initiation of subsequent anticancer therapy, whichever occurred first. Adverse events were coded using Medical Dictionary for Regulatory Activities v28.0.
[a]Combined preferred terms of Neutropenia includes neutrophil count decreased, Fatigue includes asthenia, Anemia includes hemoglobin decreased and red blood cell count decreased, and Leukopenia includes white blood cell count decreased.
ALT, alanine aminotransferase; AST, aspartate aminotransferase; IHC, immunohistochemistry; SG, sacituzumab govitecan; TEAE, treatment-emergent adverse event; TPC, treatment of physician's choice.

ASCO Annual Meeting, May 29-Jun 2, 2026, Chicago, IL, USA